RenBio®

Abstract 1101

DNA-based delivery of broadly neutralizing antibody cocktails as as universal influenza vaccine alternative

Melanie R. Walker[1], Luisa Rubinelli[1], Linda Sasset[1], Delcora Campbell[1], Diego Song[1], Andy Thompson[1], Robert Miller[1], Rachel A. Liberatore[1], Marek M. Drozdz[1]

[1]RenBio, Long Island City, New York, USA; email: *rachel.liberatore@renbio.com, marek.drozdz@renbio.com*

MYO Technology Platform

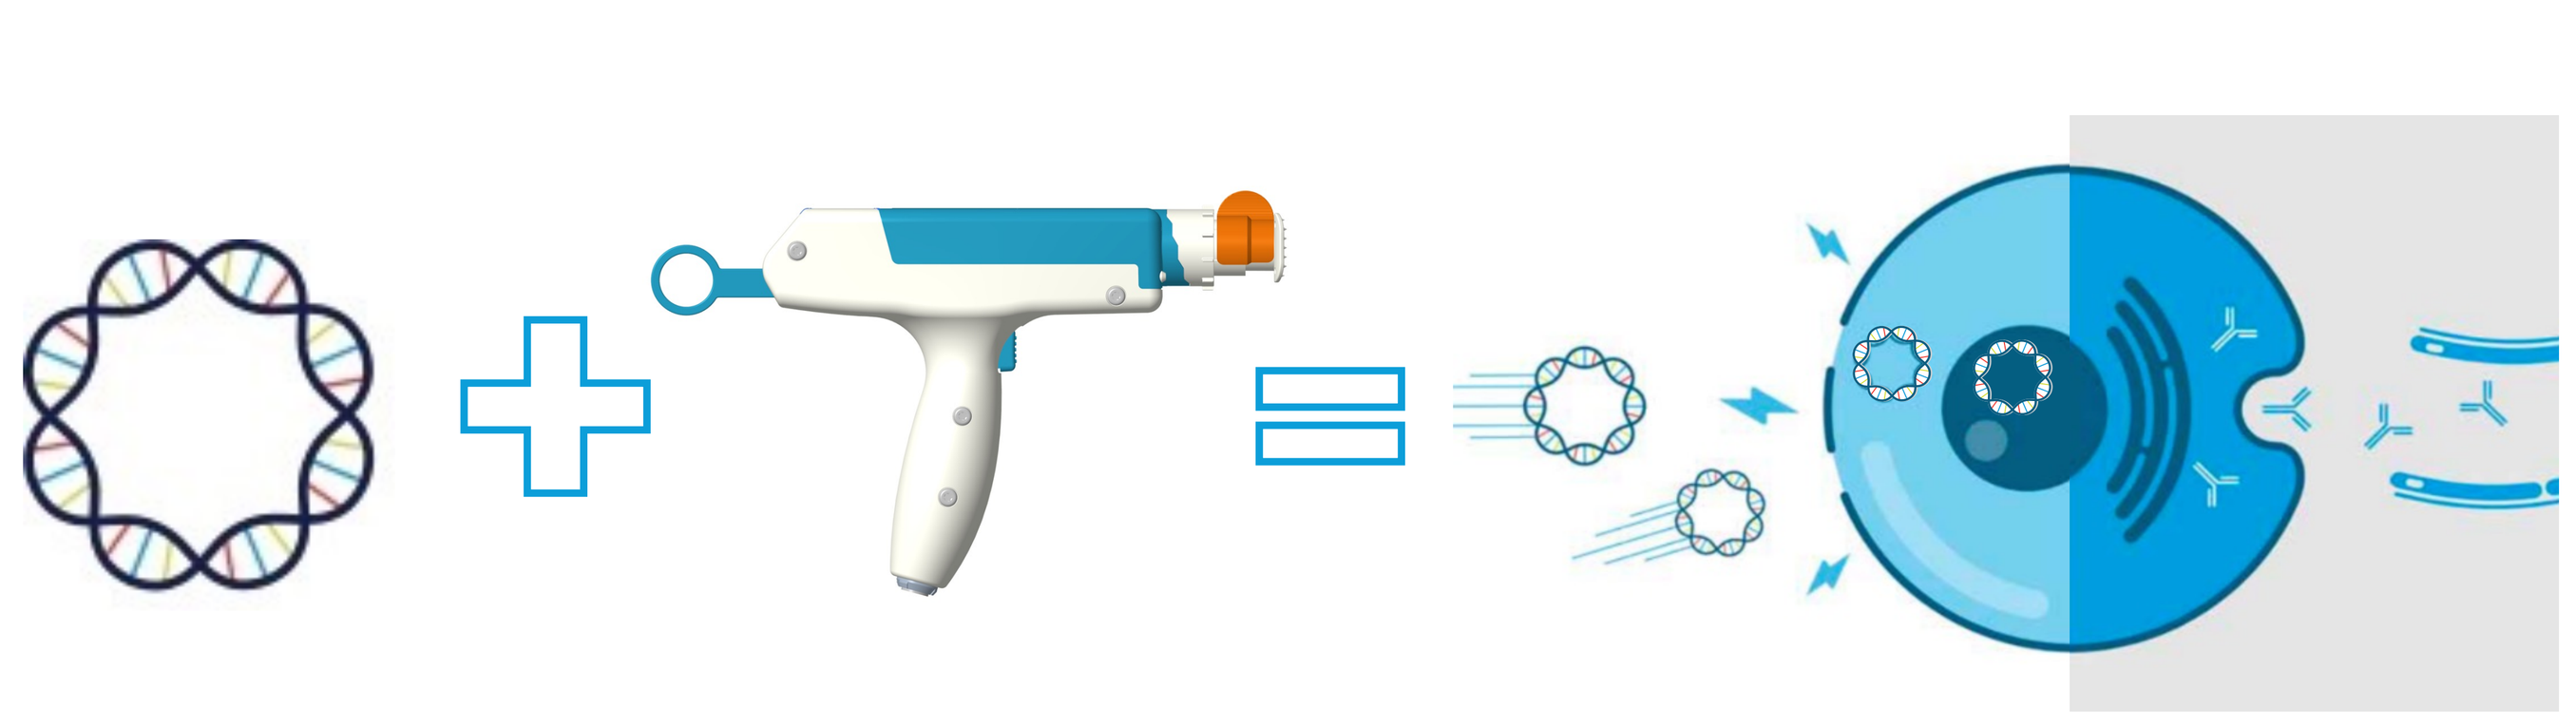

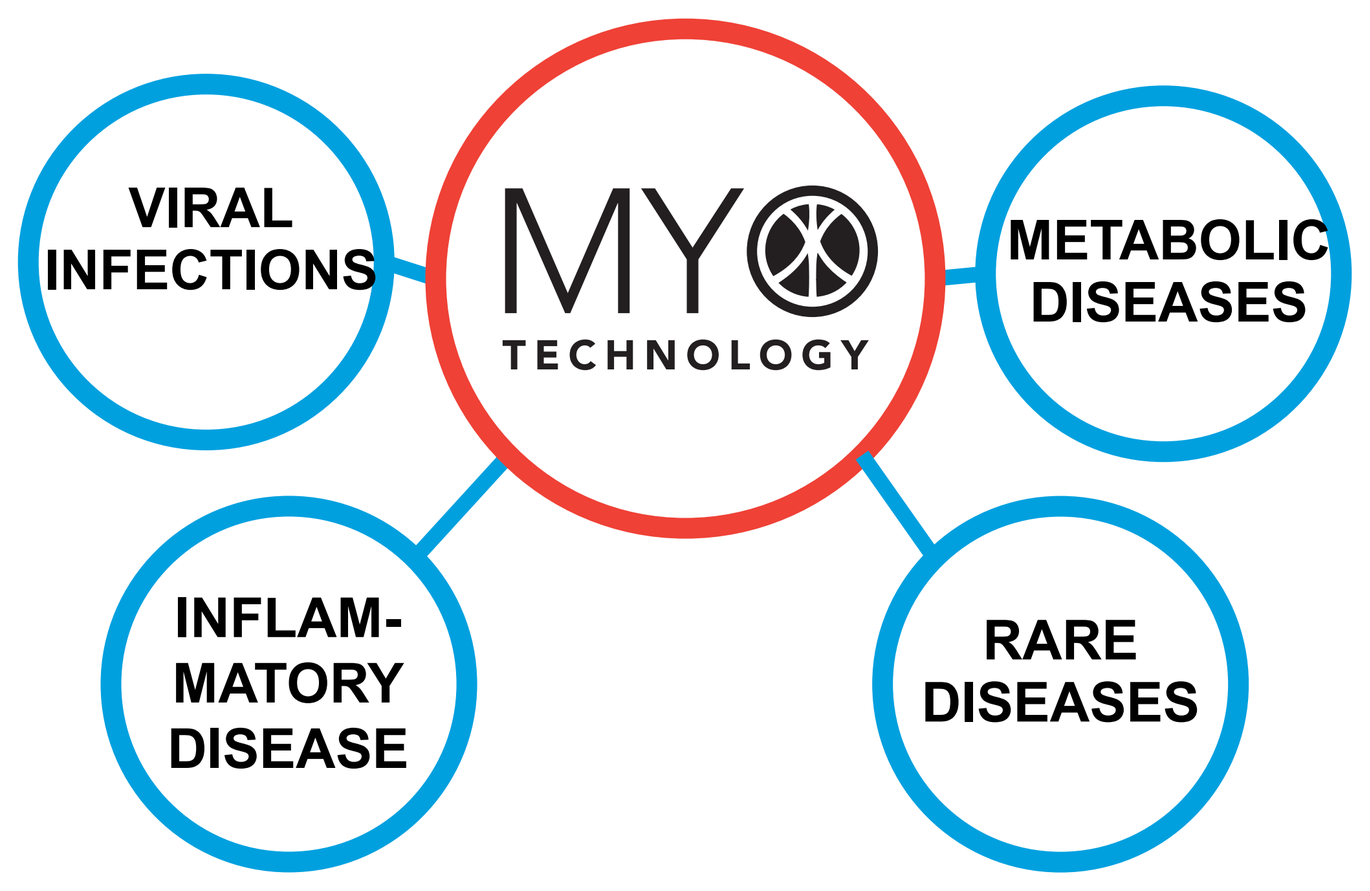
VIRAL INFECTIONS
MYO TECHNOLOGY
METABOLIC DISEASES
INFLAM-MATORY DISEASE
RARE DISEASES

The MYO Technology™ platform enables ultra-long-lasting delivery of antibody and protein therapeutics via intramuscular electroporation (EP) of plasmid DNA. Transfected muscle cells durably express and secrete the encoded protein, which is taken up into peripheral circulation. This uniquely extended pharmacokinetic profile is ideal for treatment of chronic diseases, and prevention of infectious disease.

MYO delivery of antibody genes results in durable antibody levels *in vivo*

Traditional antibody protein delivery

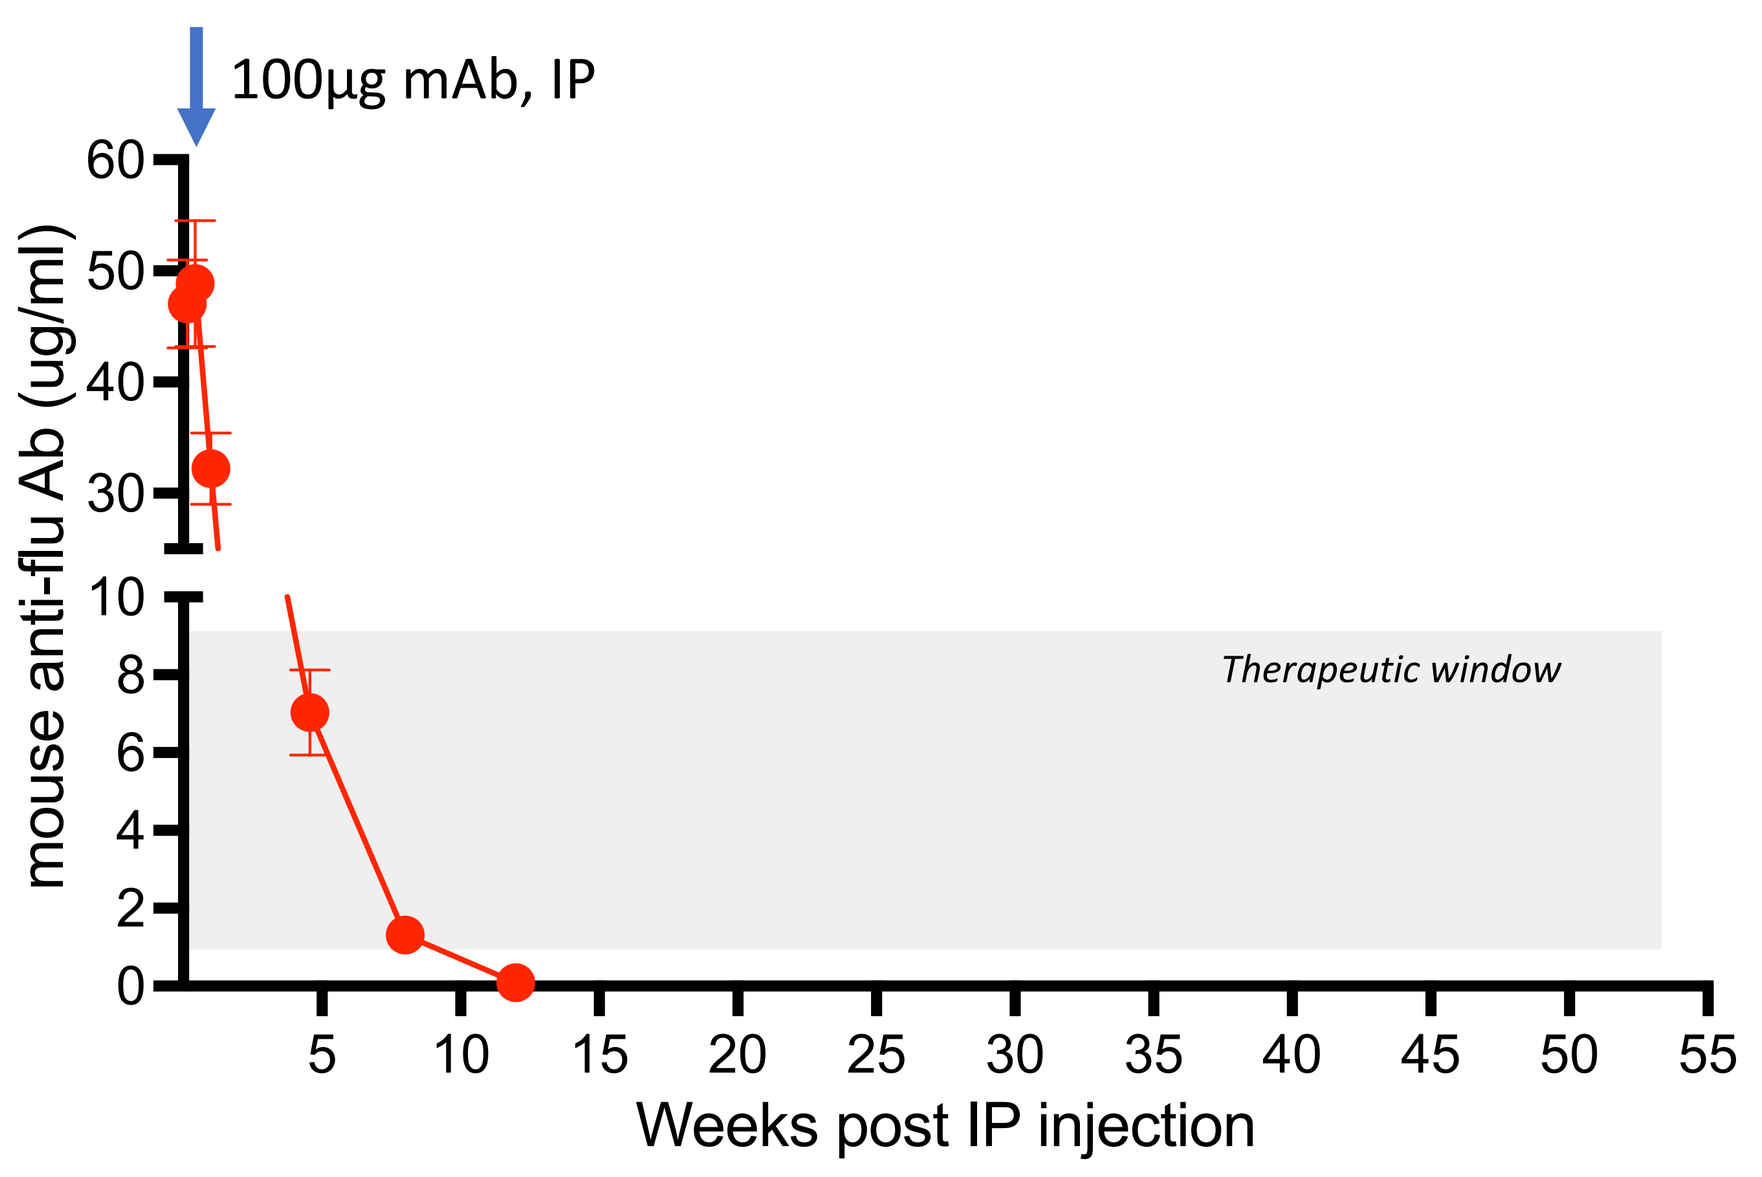
100µg mAb, IP
mouse anti-flu Ab (ug/ml)
Therapeutic window
Weeks post IP injection

MYO delivery of antibody genes

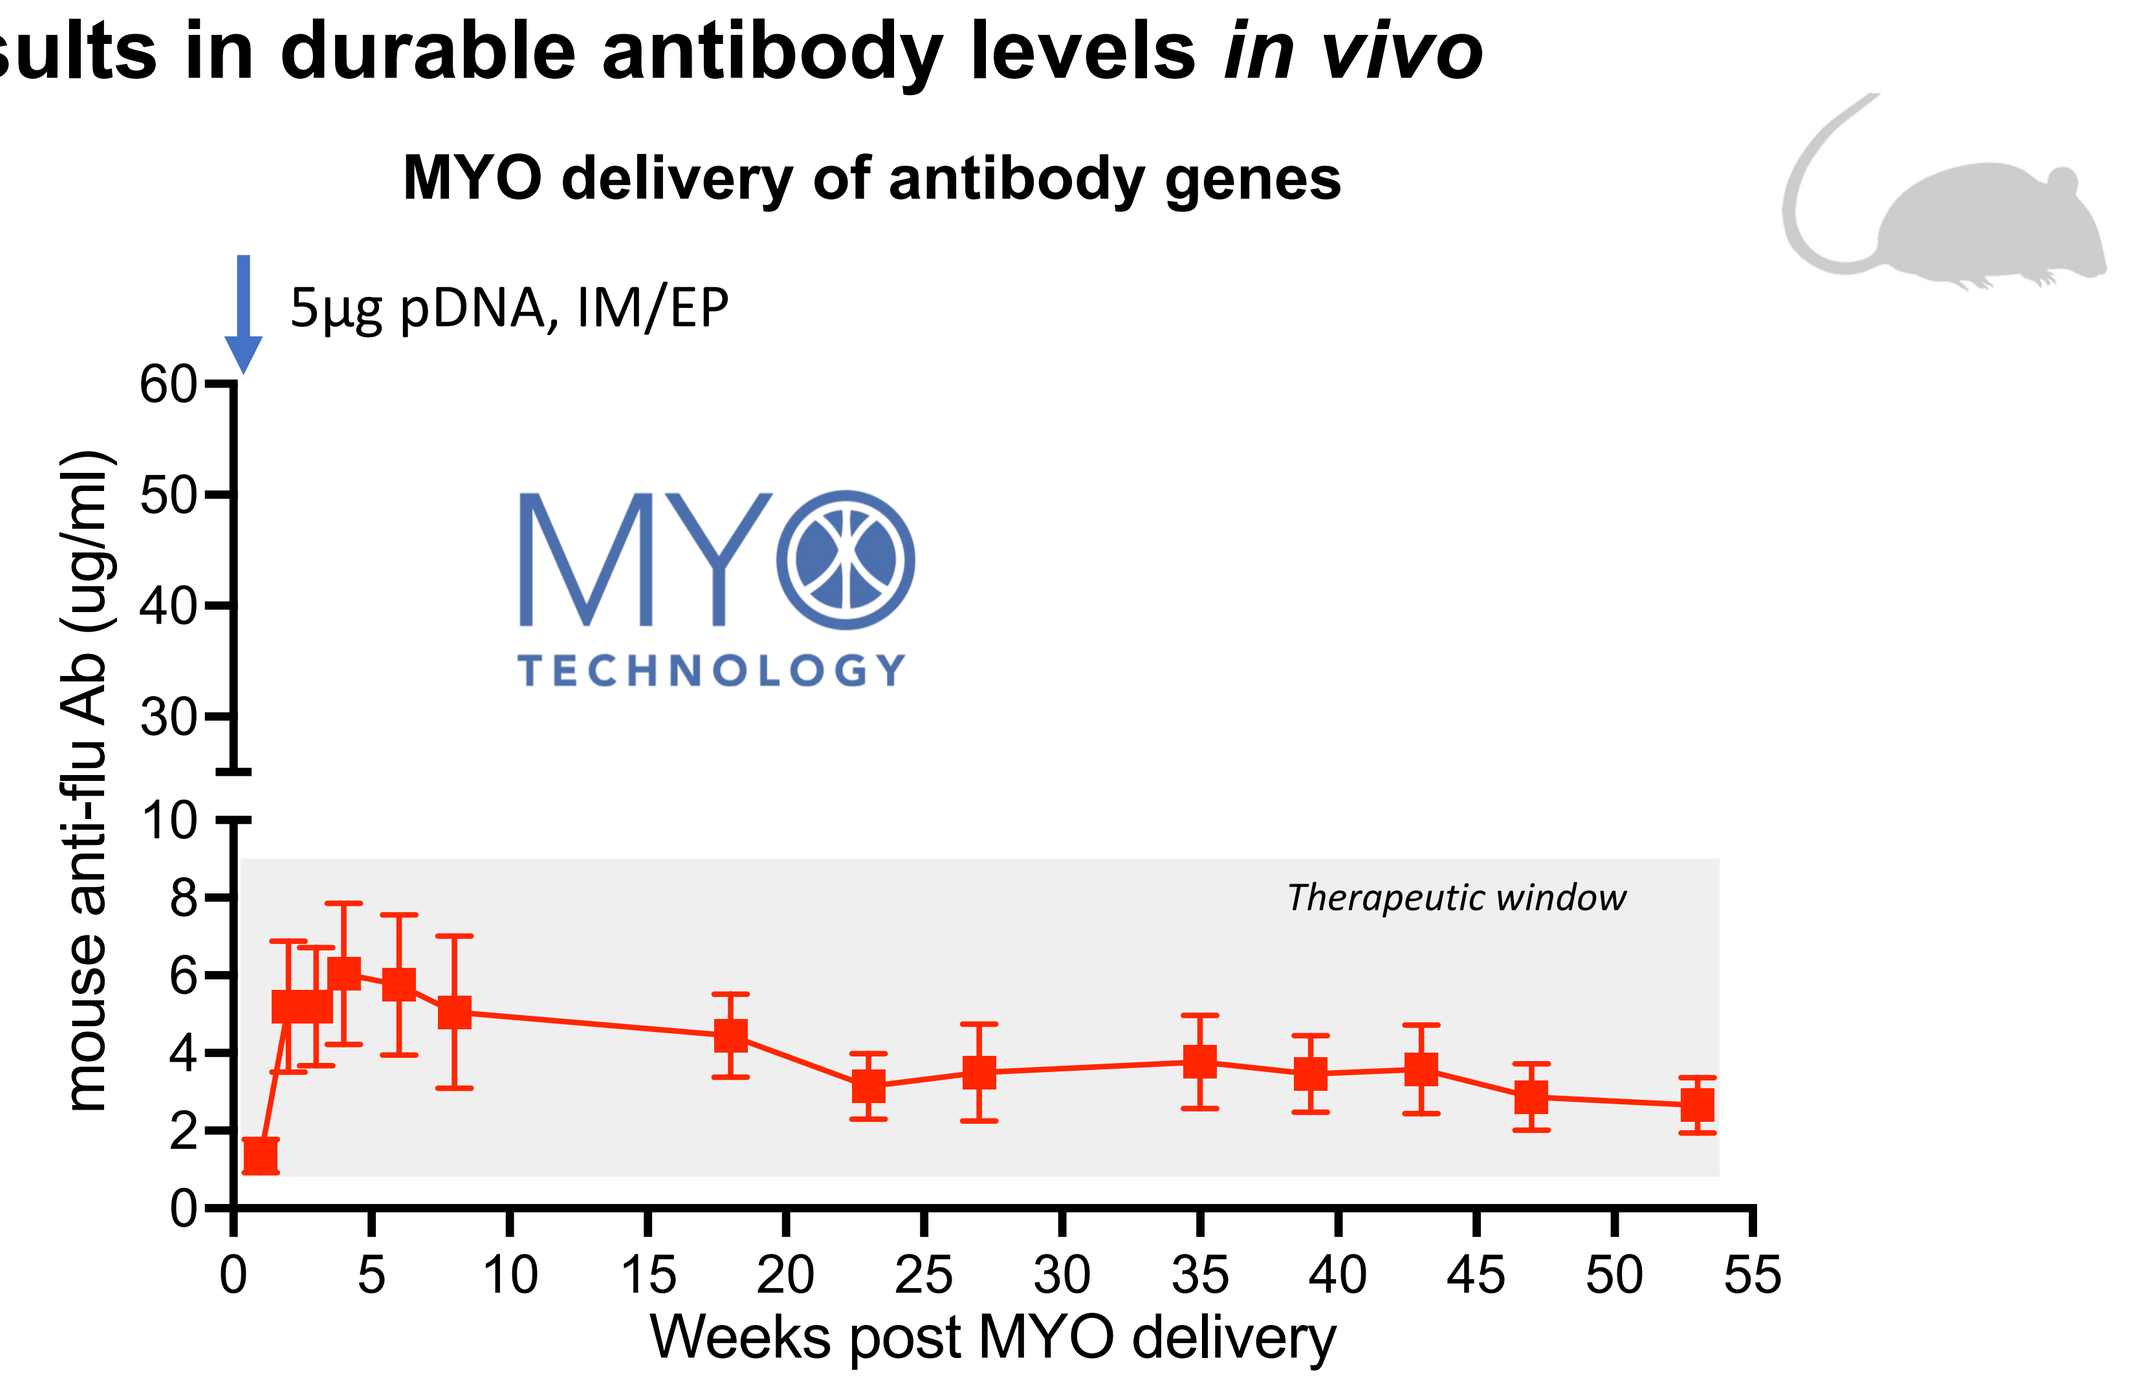
5µg pDNA, IM/EP
MYO TECHNOLOGY
mouse anti-flu Ab (ug/ml)
Therapeutic window
Weeks post MYO delivery

Serum antibody titration with pDNA dose escalation

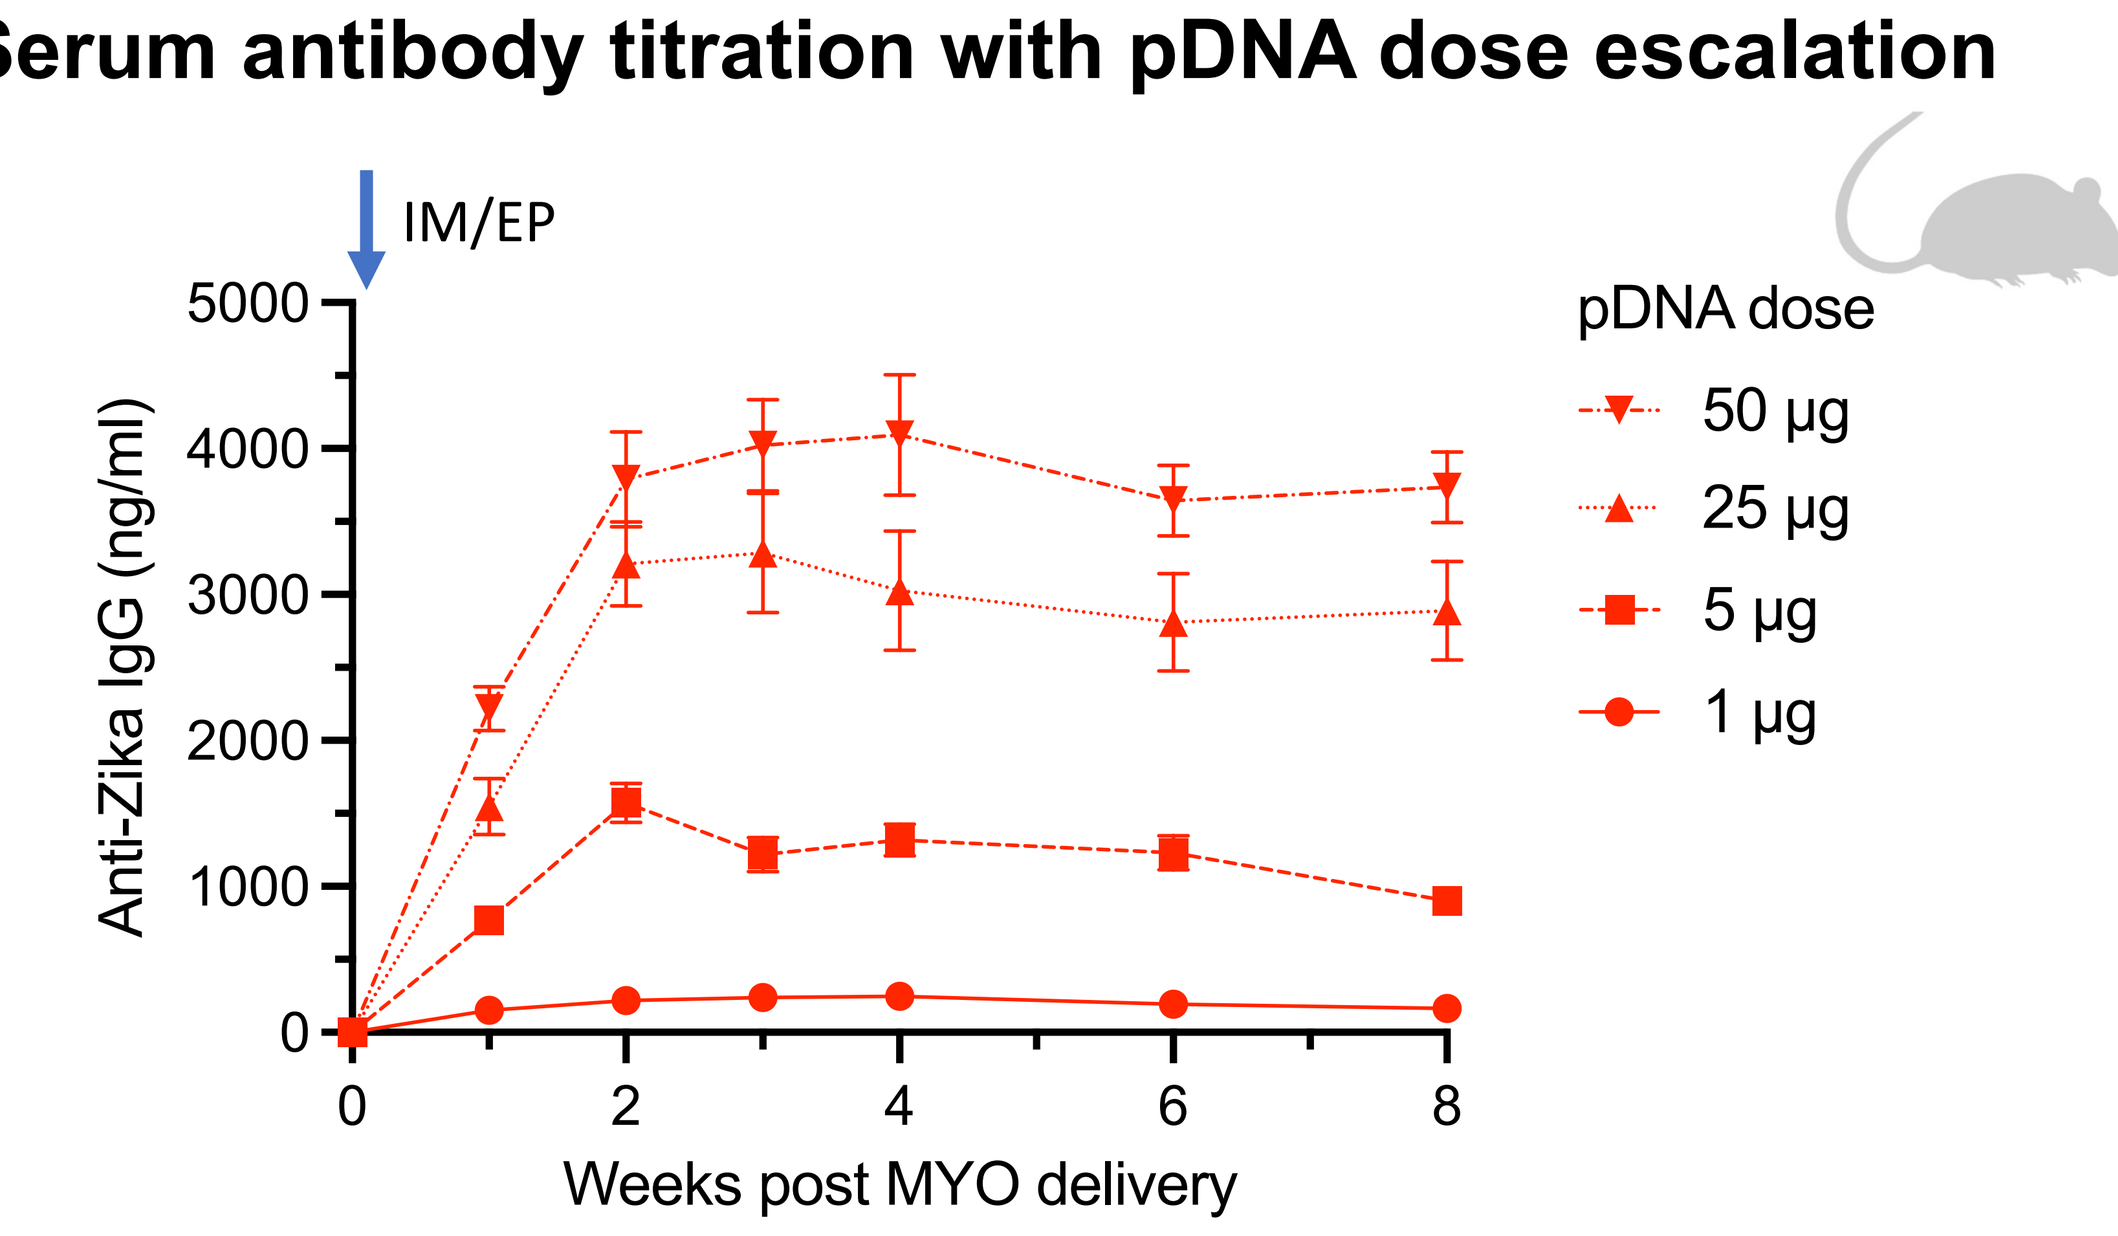
IM/EP
Anti-Zika IgG (ng/ml)
pDNA dose
50 µg
25 µg
5 µg
1 µg
Weeks post MYO delivery

Evaluation of durability and scale with species-matched mAbs in rabbits

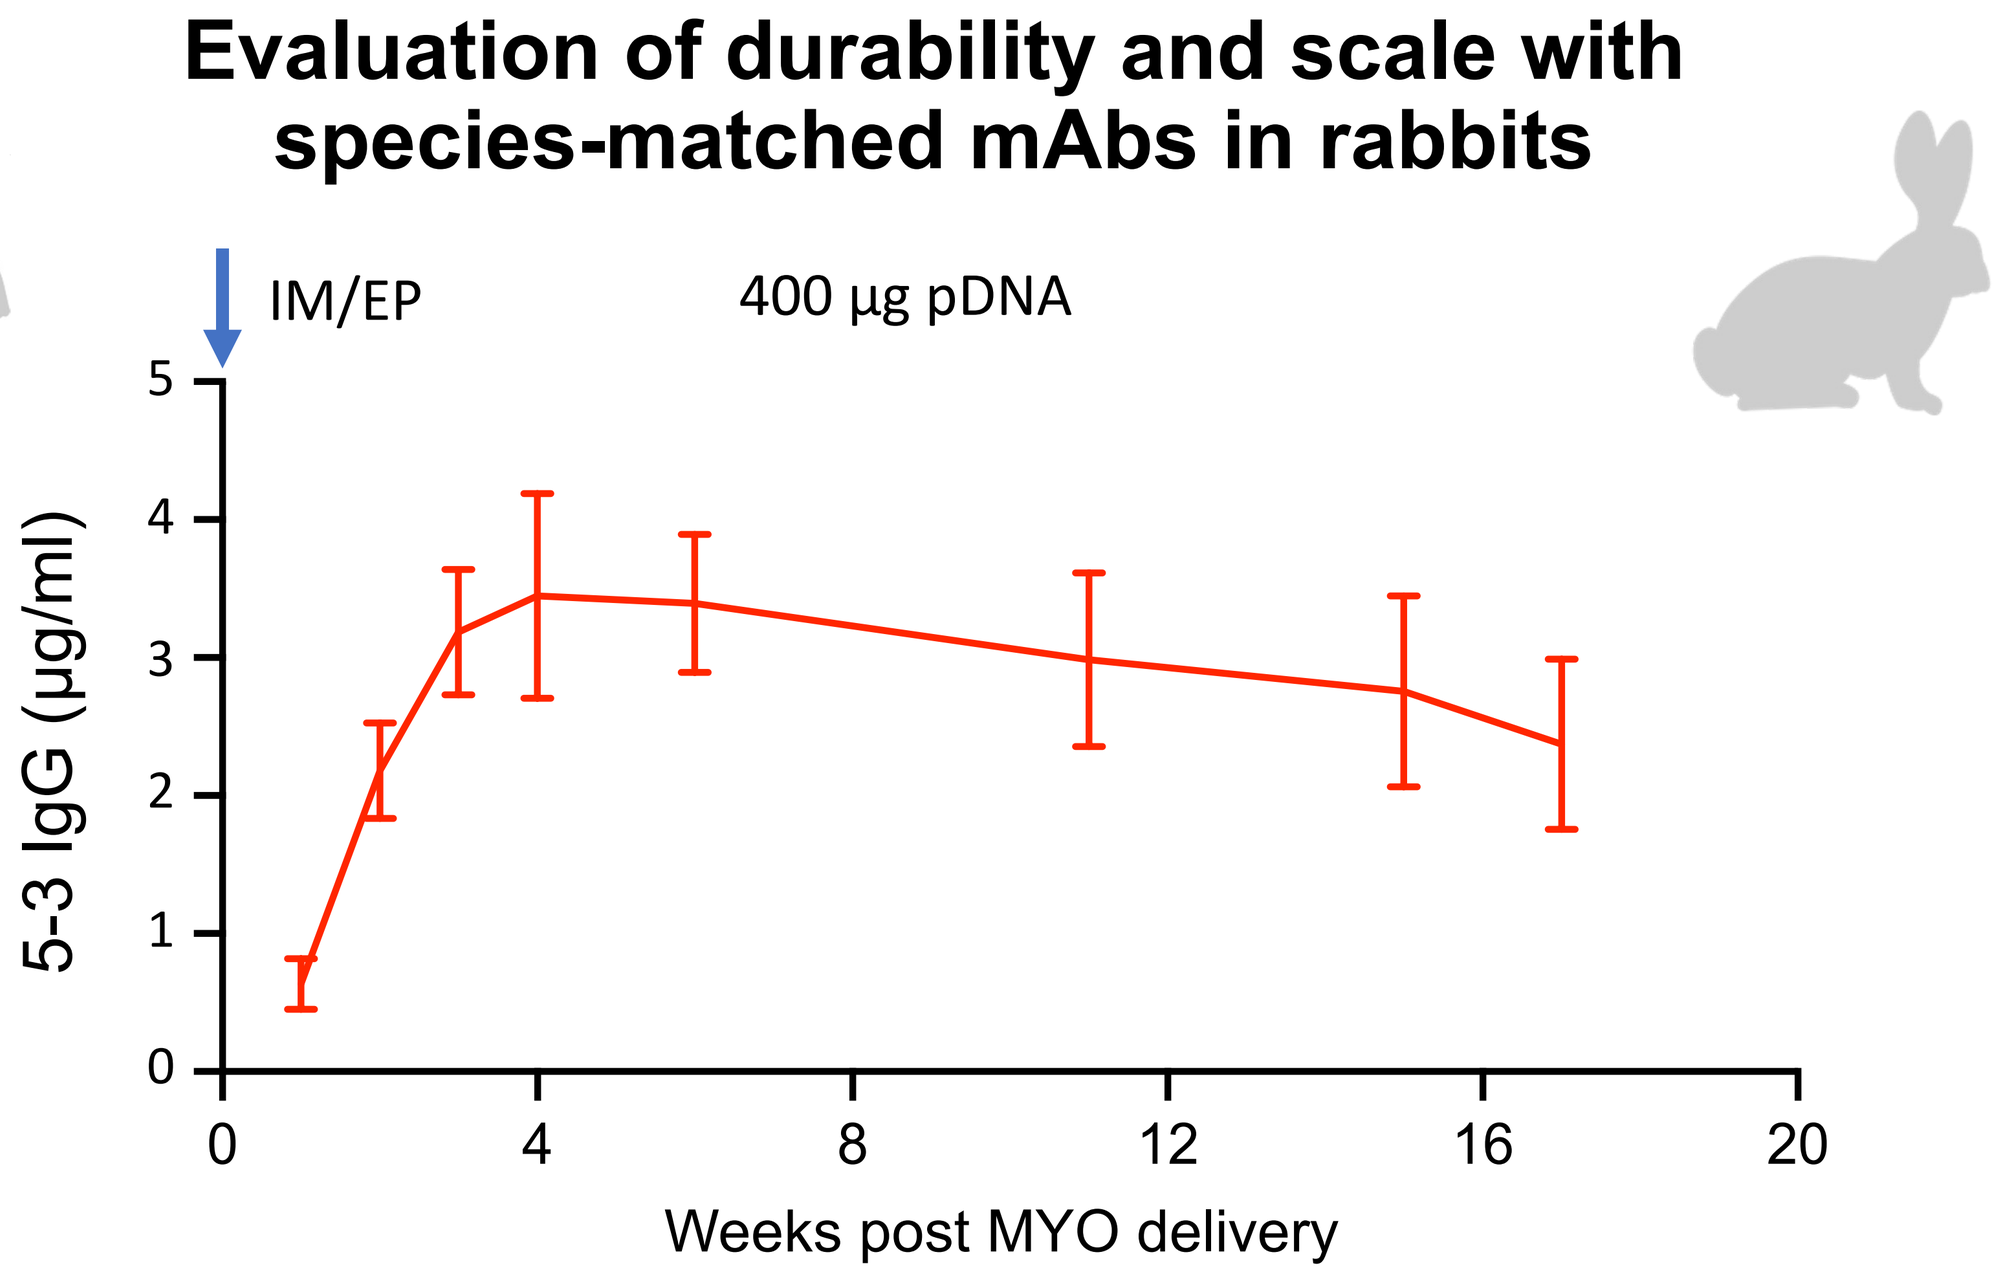
IM/EP
400 µg pDNA
5-3 IgG (µg/ml)
Weeks post MYO delivery

Efficacy Data in Disease Models

Neutropenia

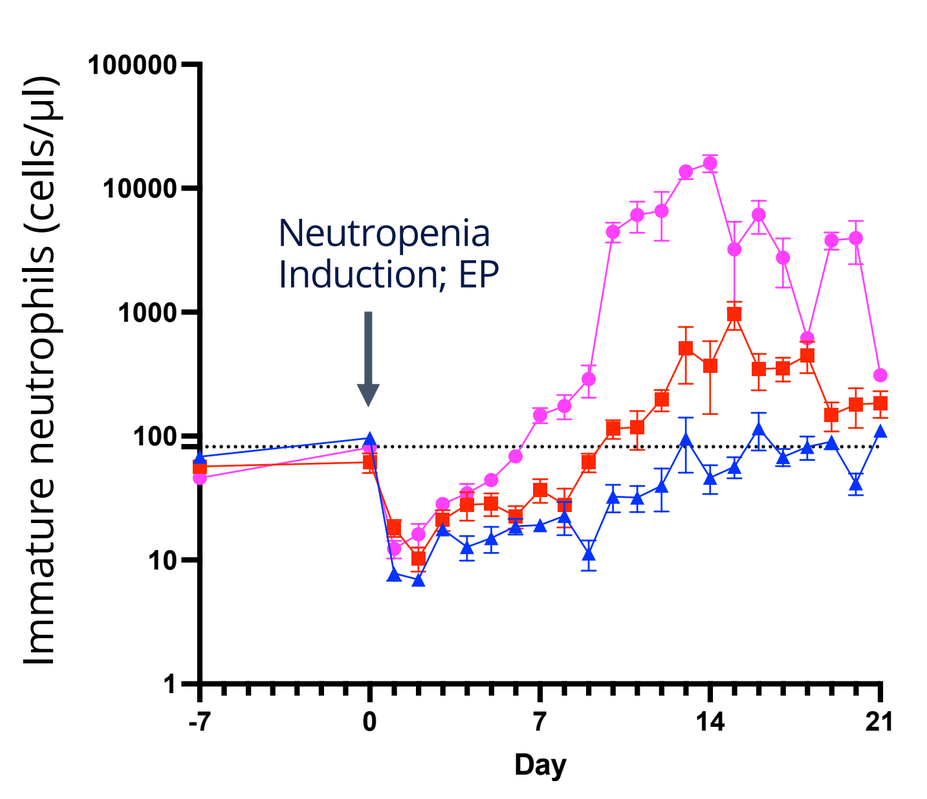
Immature neutrophils (cells/µl)
Neutropenia Induction; EP
Untreated
1ug G-CSF
5ug G-CSF
Day

Drozdz MM, et al. Molecular Therapy (2025), https://doi.org/10.1016/j.ymthe.2025.03.038

Psoriasis

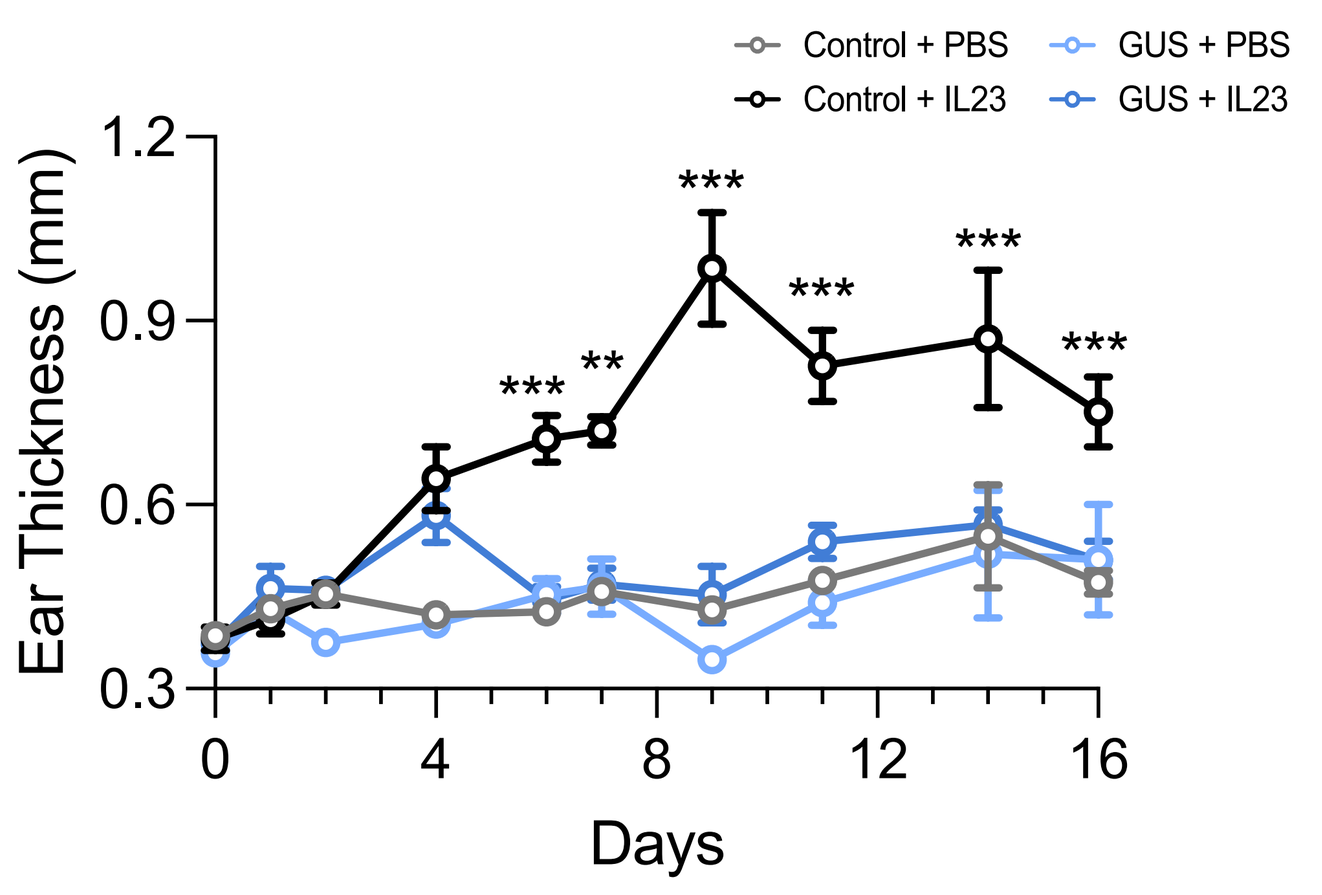
Control + PBS
Control + IL23
GUS + PBS
GUS + IL23
Ear Thickness (mm)
Days

Influenza

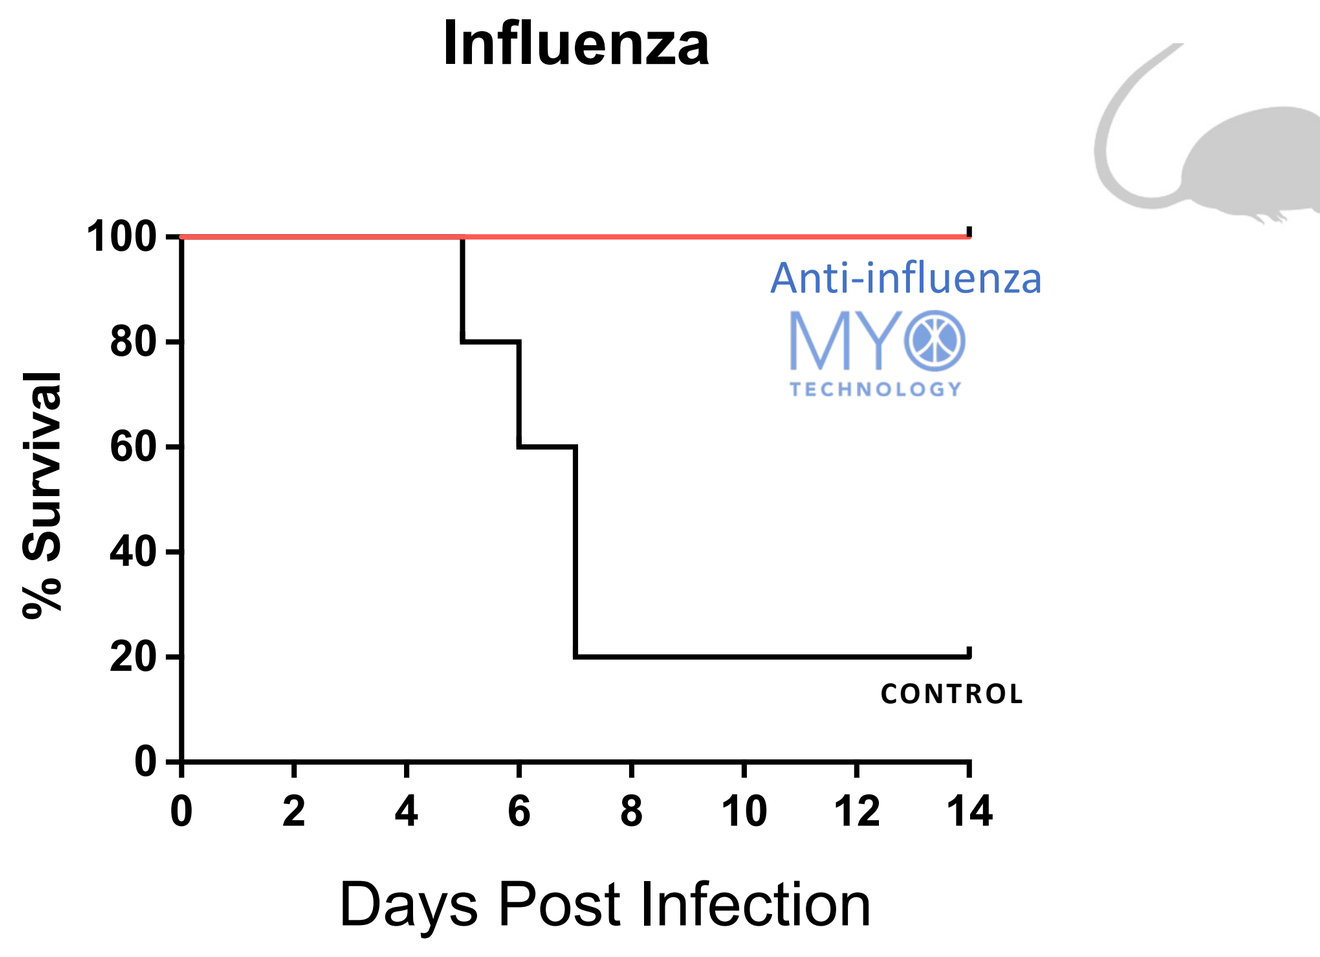
% Survival
Anti-influenza MYO TECHNOLOGY
CONTROL
Days Post Infection

Obesity

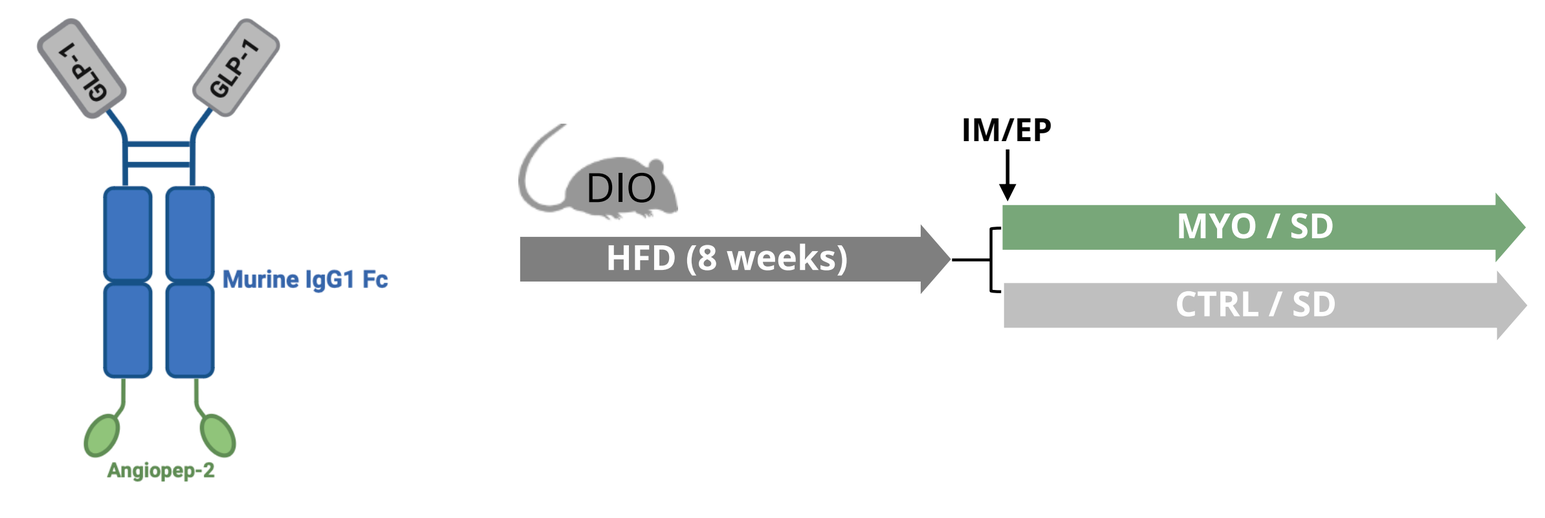
GLP-1
GLP-1
Murine IgG1 Fc
Angiopep-2
DIO
HFD (8 weeks)
IM/EP
MYO / SD
CTRL / SD

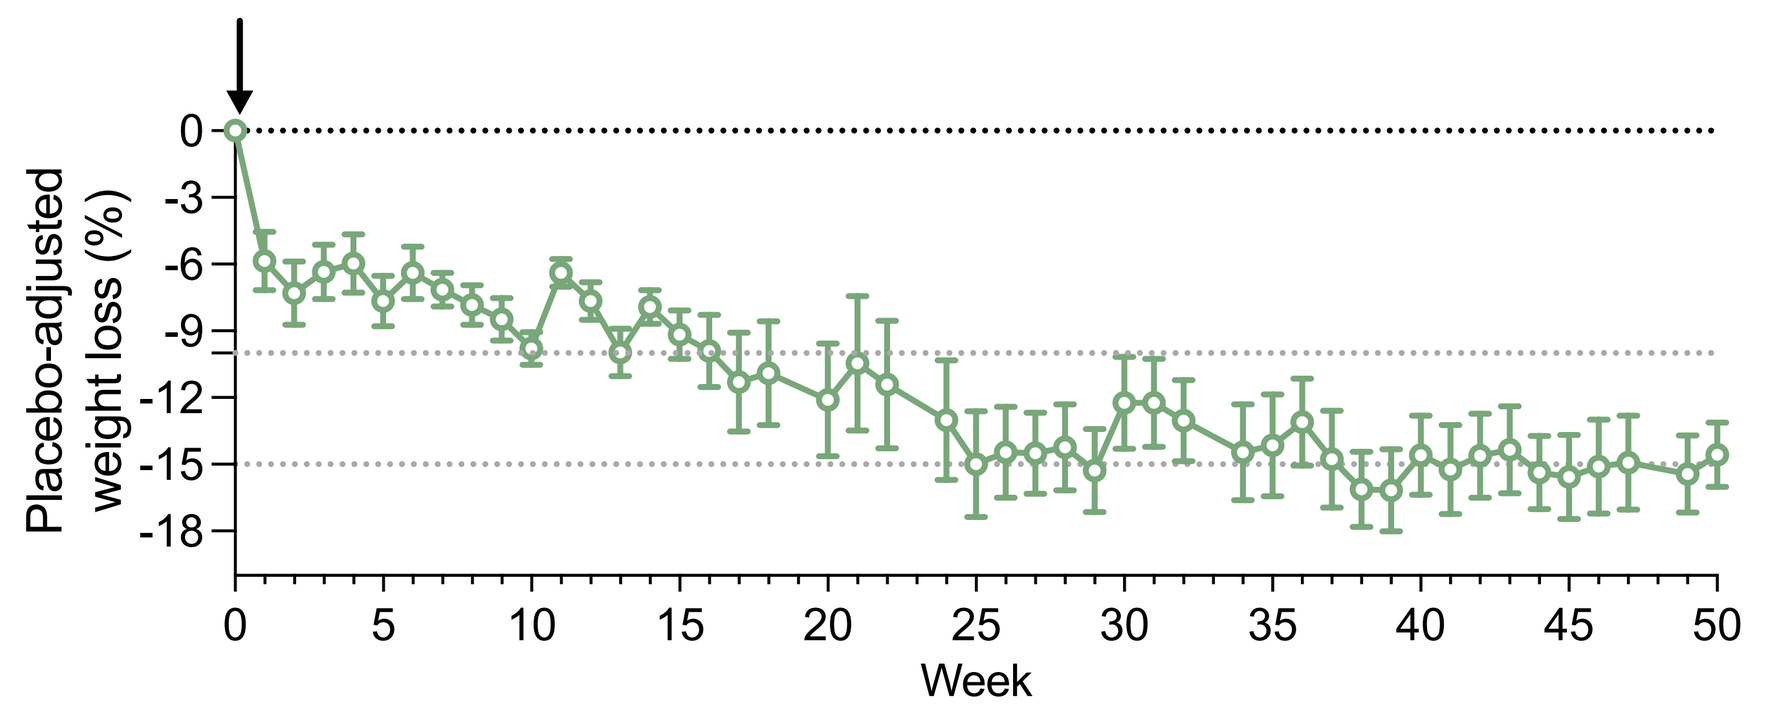
Placebo-adjusted weight loss (%)
Week

In vivo-produced antibodies and therapeutic proteins are functional and efficacious in a diverse array of disease models.

Neutralizing Antibody Cocktail for Influenza Prevention

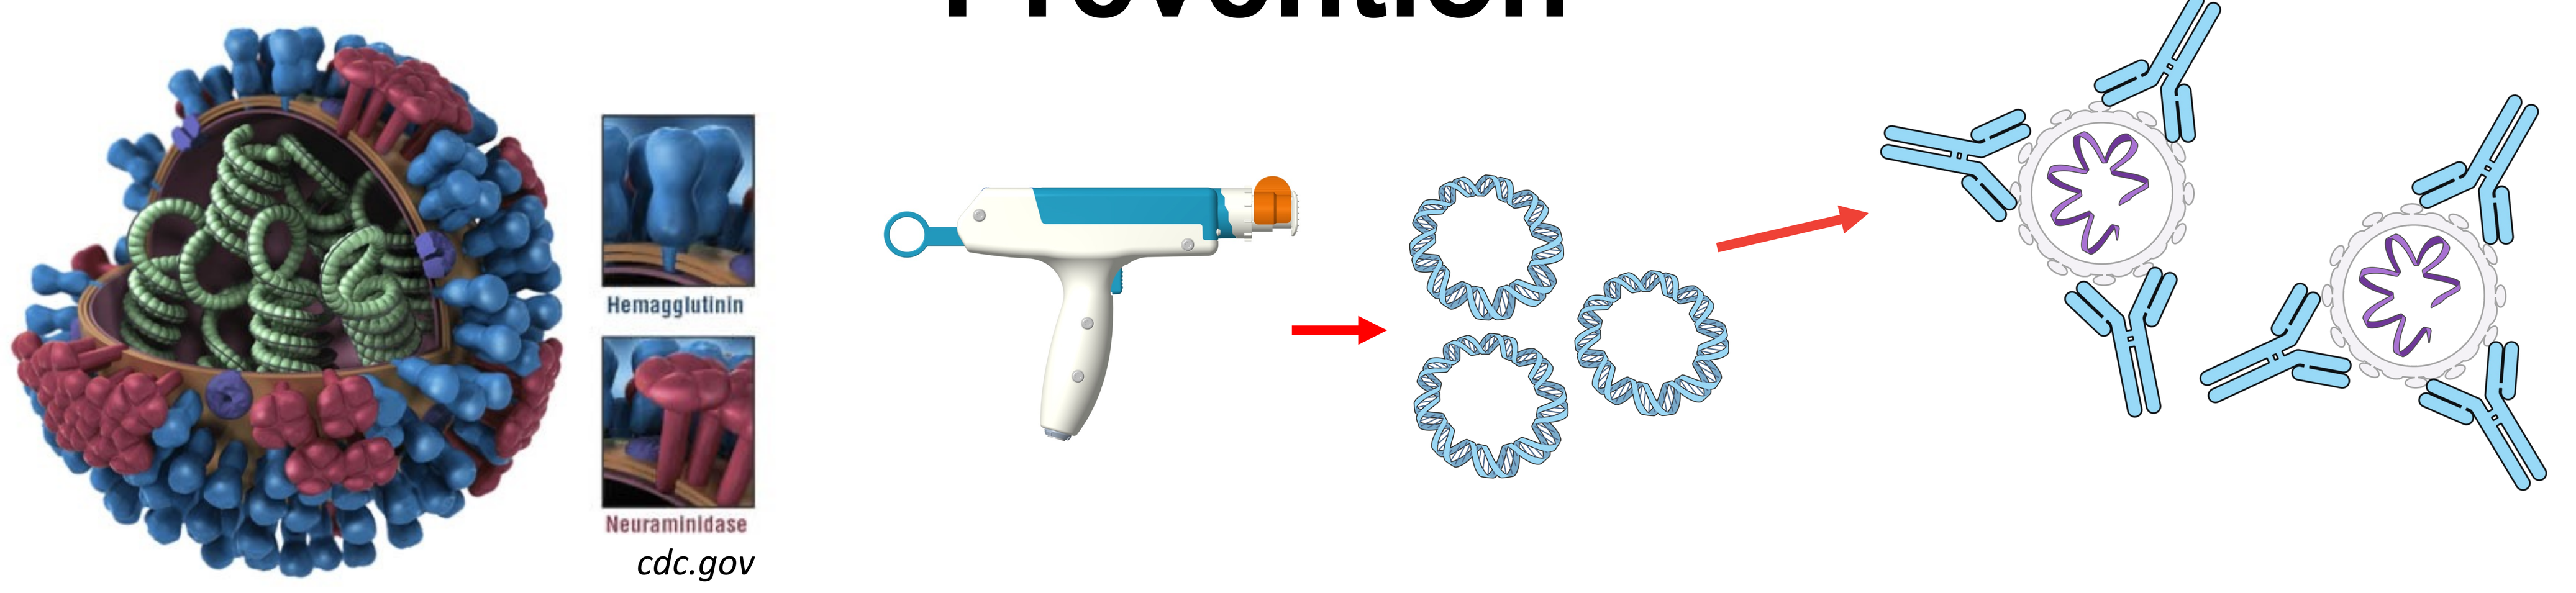
Hemagglutinin
Neuraminidase
cdc.gov

Neutralizing antibodies (NAbs) against influenza viruses can prevent disease in preclinical and clinical settings. We hypothesized that pDNA-based delivery of NAb cocktails using MYO Technology could offer significant and extended protection against a broad range of influenza viruses by targeting conserved regions on antigenic proteins.

NAb Cocktail Delivery via Ab Engineering

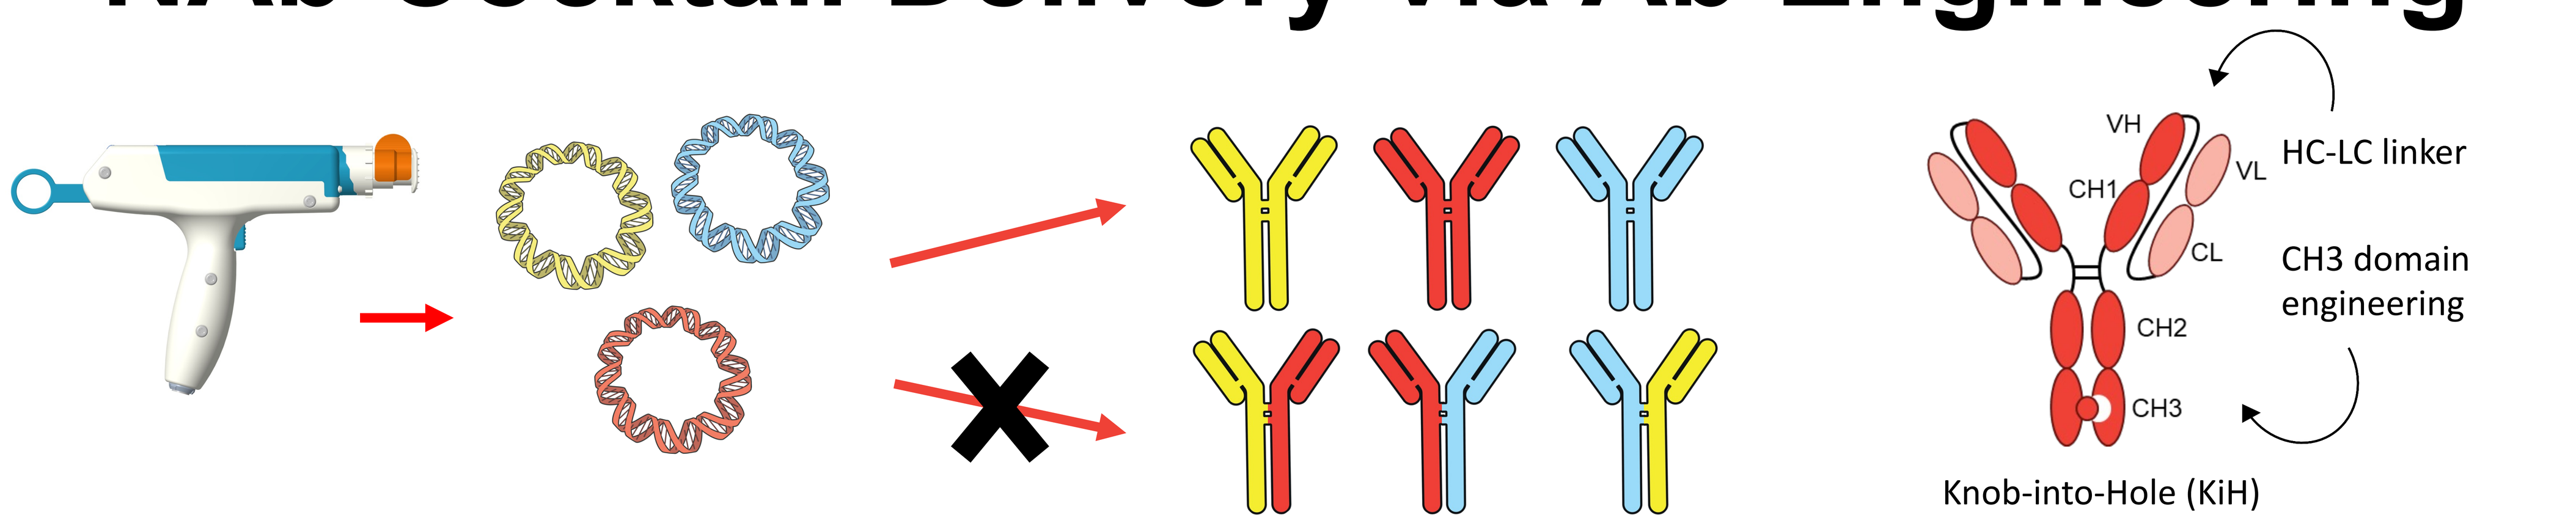
VH
VL
CH1
CL
CH2
CH3
HC-LC linker
CH3 domain engineering
Knob-into-Hole (KiH)

Antibody engineering techniques ensure proper antibody assembly and prevent the formation of mispaired antibody hybrids *in vivo.*

Fc engineering promotes stable antibody formation *in vivo* post EP

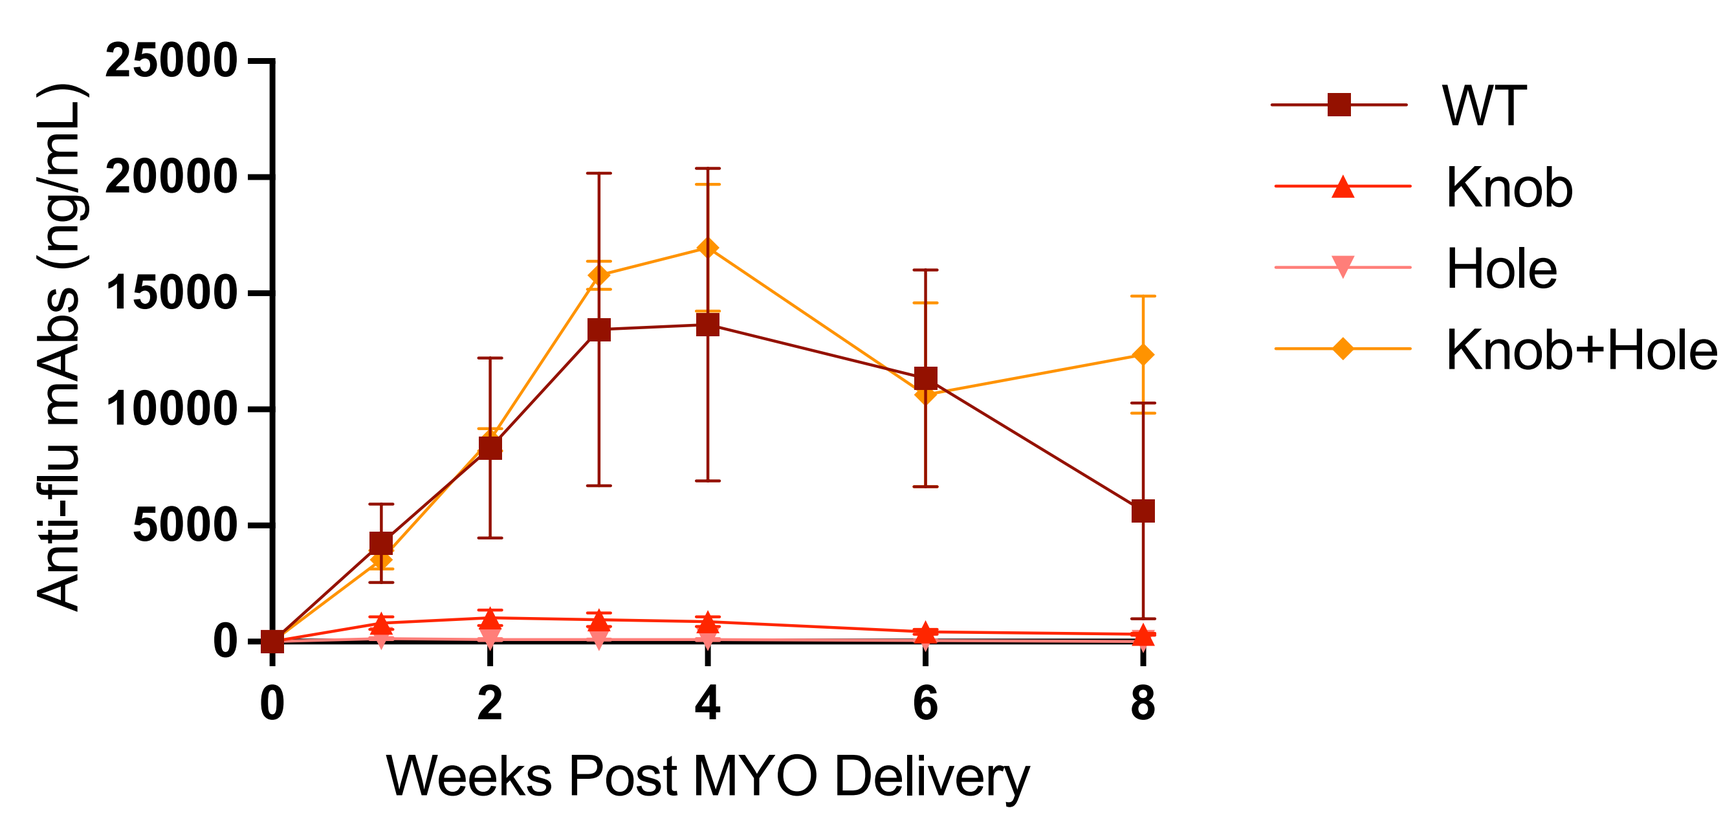
Anti-flu mAbs (ng/mL)
WT
Knob
Hole
Knob+Hole
Weeks Post MYO Delivery

Fc engineering reduces formation of mispaired antibody hybrids

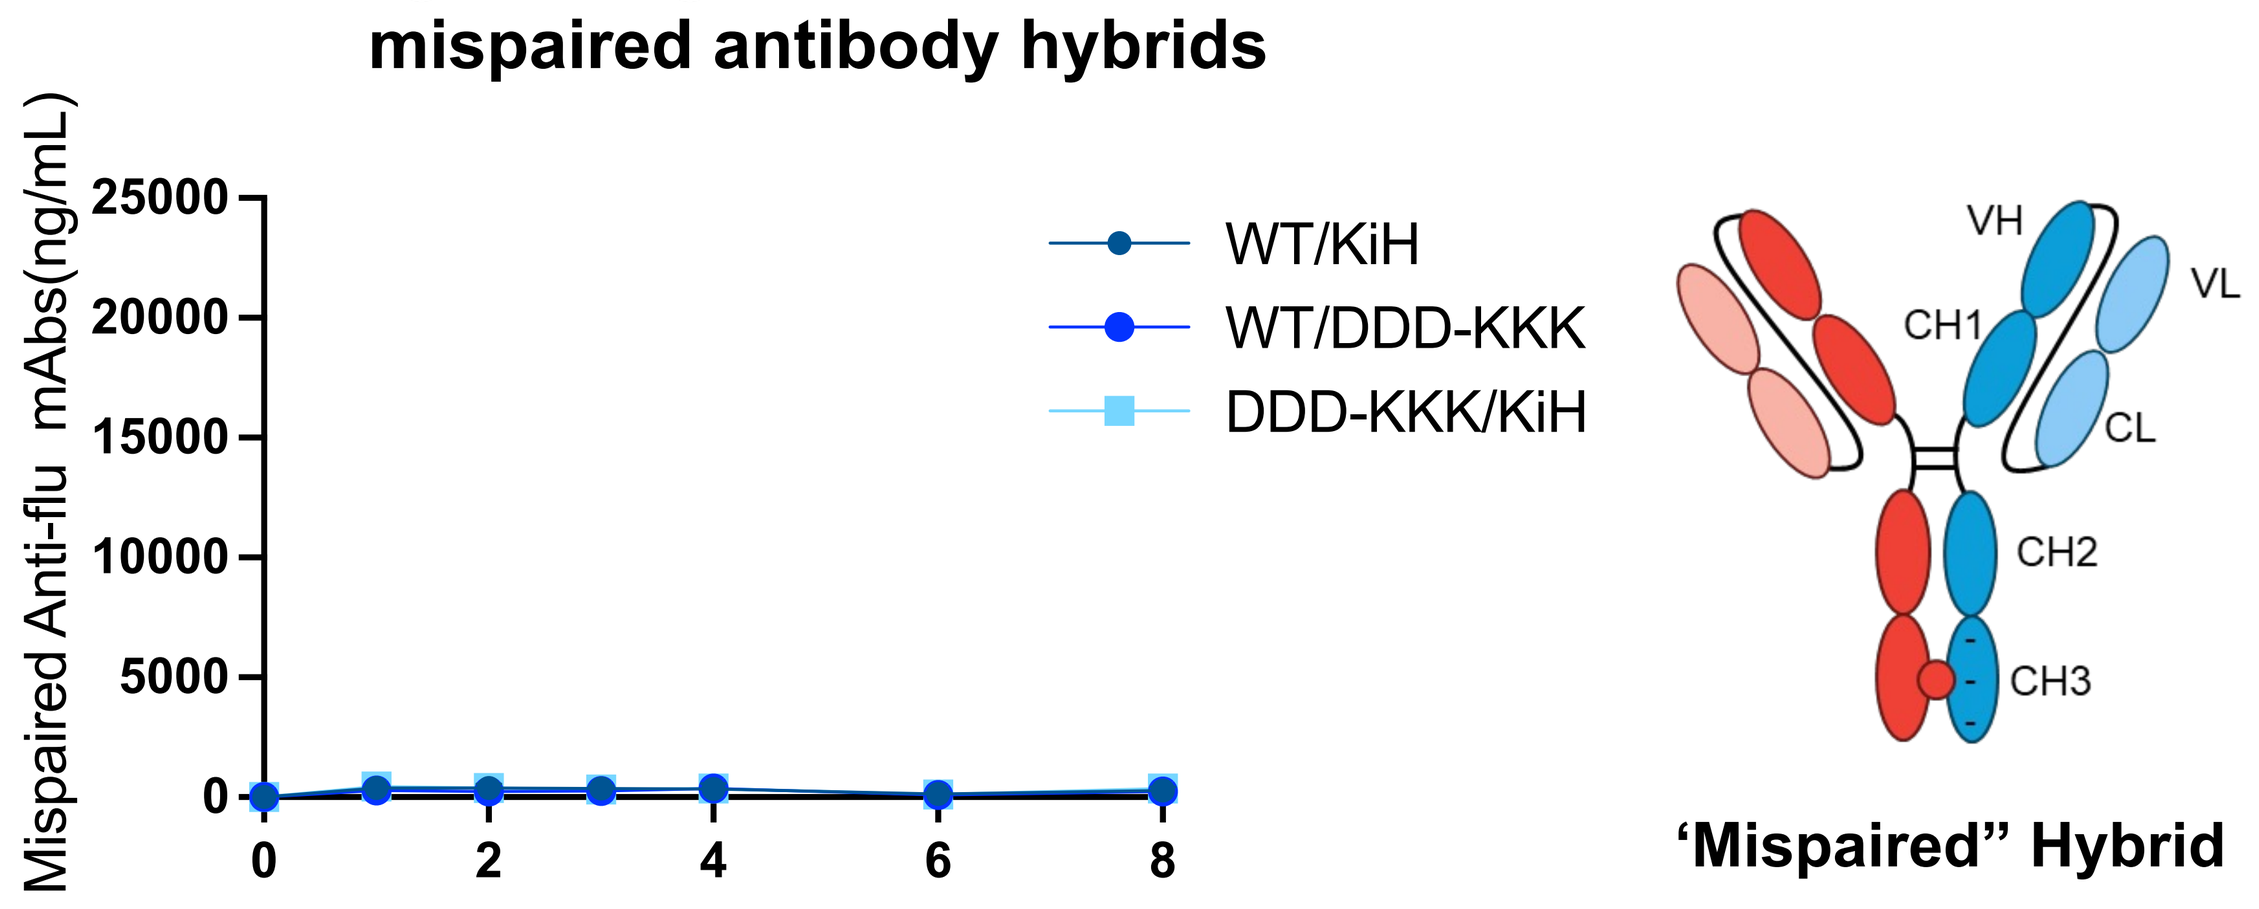
Mispaired Anti-flu mAbs(ng/mL)
WT/KiH
WT/DDD-KKK
DDD-KKK/KiH
Weeks Post MYO Delivery
VH
VL
CH1
CL
CH2
CH3
'Mispaired" Hybrid

Cocktail	Antibody #1	Antibody #2	Antibody #3
1	CR9114 (HA stem)	5J8 (head – H1)	F045-092 (head – H3)
2	FNI9 (NA)	5J8 (head – H1)	F045-092 (head – H3)
3	2-12C (head – H1)	5J8 (head – H1)	F045-092 (head – H3)
4	K03.12 (head – H3)	5J8 (head – H1)	F045-092 (head – H3)
Mod/Tag	WT – Flag	DDD-KKK – His	KiH – Myc

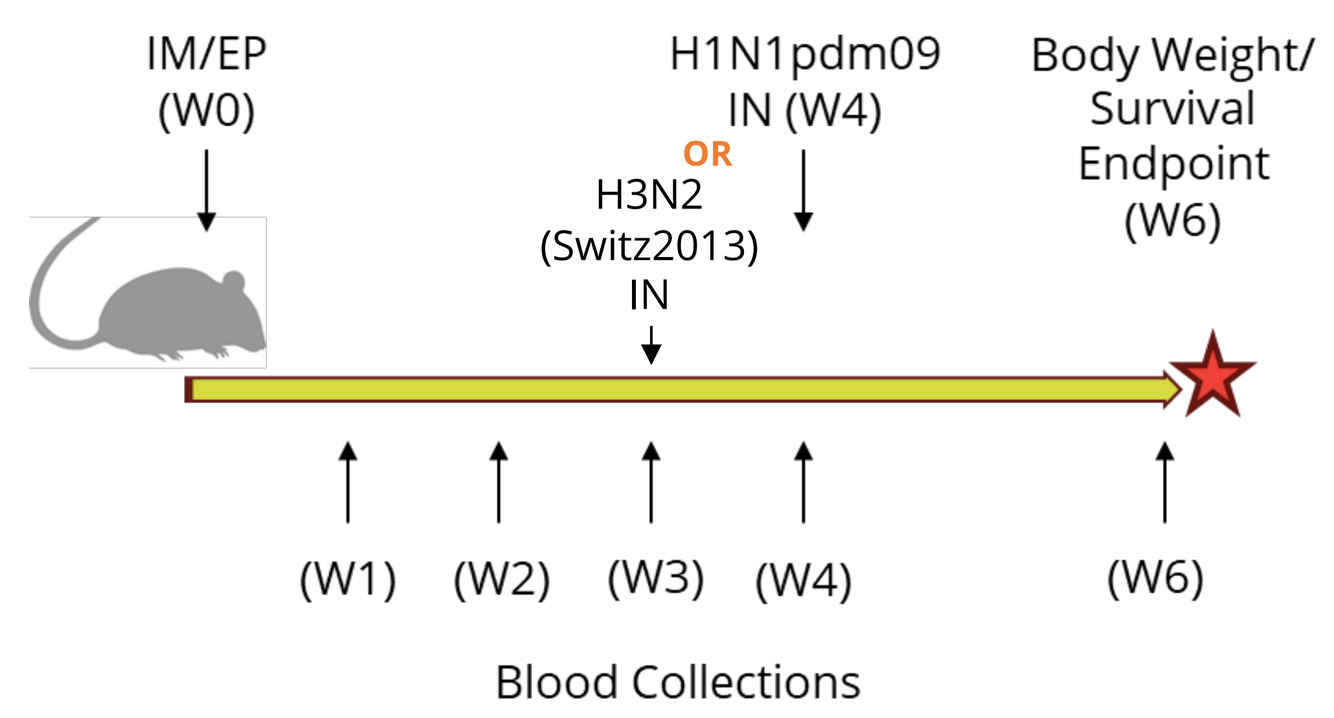
IM/EP (W0)
H1N1pdm09 IN (W4)
OR
H3N2 (Switz2013) IN
Body Weight/ Survival Endpoint (W6)
(W1)
(W2)
(W3)
(W4)
(W6)
Blood Collections

A single cocktail can protect against both H1N1 and H3N2 viruses

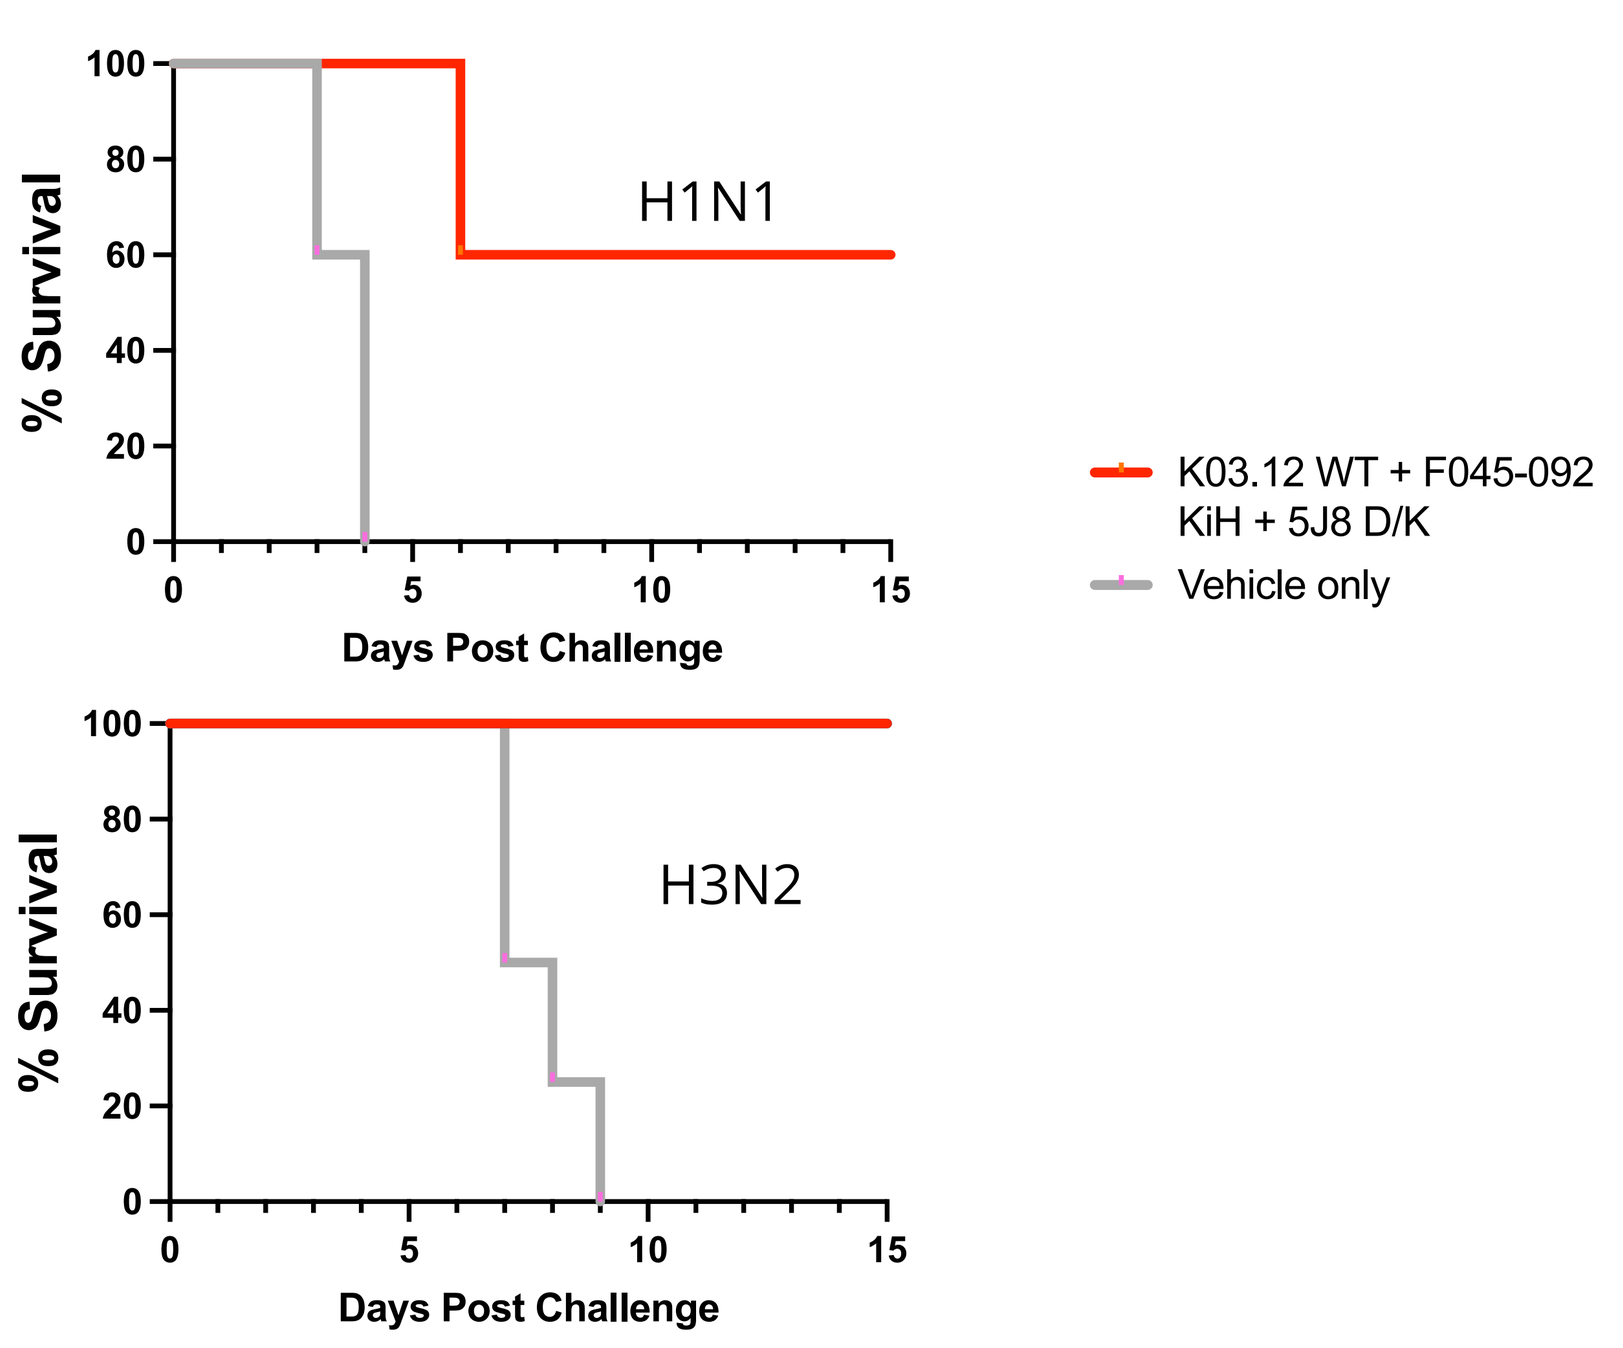
% Survival
H1N1
Days Post Challenge
K03.12 WT + F045-092 KiH + 5J8 D/K
Vehicle only
H3N2
Days Post Challenge

[2] Gunasekaran et al, J Biol Chem. 2010 Jun 18;285(25):19637-46. doi: 10.1074/jbc.M110.117382.

Acknowledgements

wellcome
BARDA
New York State of Opportunity. Empire State Development
DARPA
Bill & Melinda Gates foundation